PAN-CASPASE INHIBITION PROTECTS AGAINST FIBROTIC NASH INDUCED BY CHOLINE DEFICIENT AMINO ACID DEFINED DIET (CDAA)

W. Lu[1], A. Eguchi[1], D. Sirbu[1], P. Contreras[2], C. Johnson[1], A. Wree[1], D. Povero[1], M. Lazic[1], and A. Feldstein[1]
[1]*Pediatrics, University of California, San Diego, La Jolla,* [2]*Conatus Pharmaceuticals, San Diego, CA, USA.*

UC San Diego MEDICAL CENTER

PURPOSE / AIM

- ➢ Hepatocyte cell death is a key feature of nonalcoholic steatohepatitis (NASH)

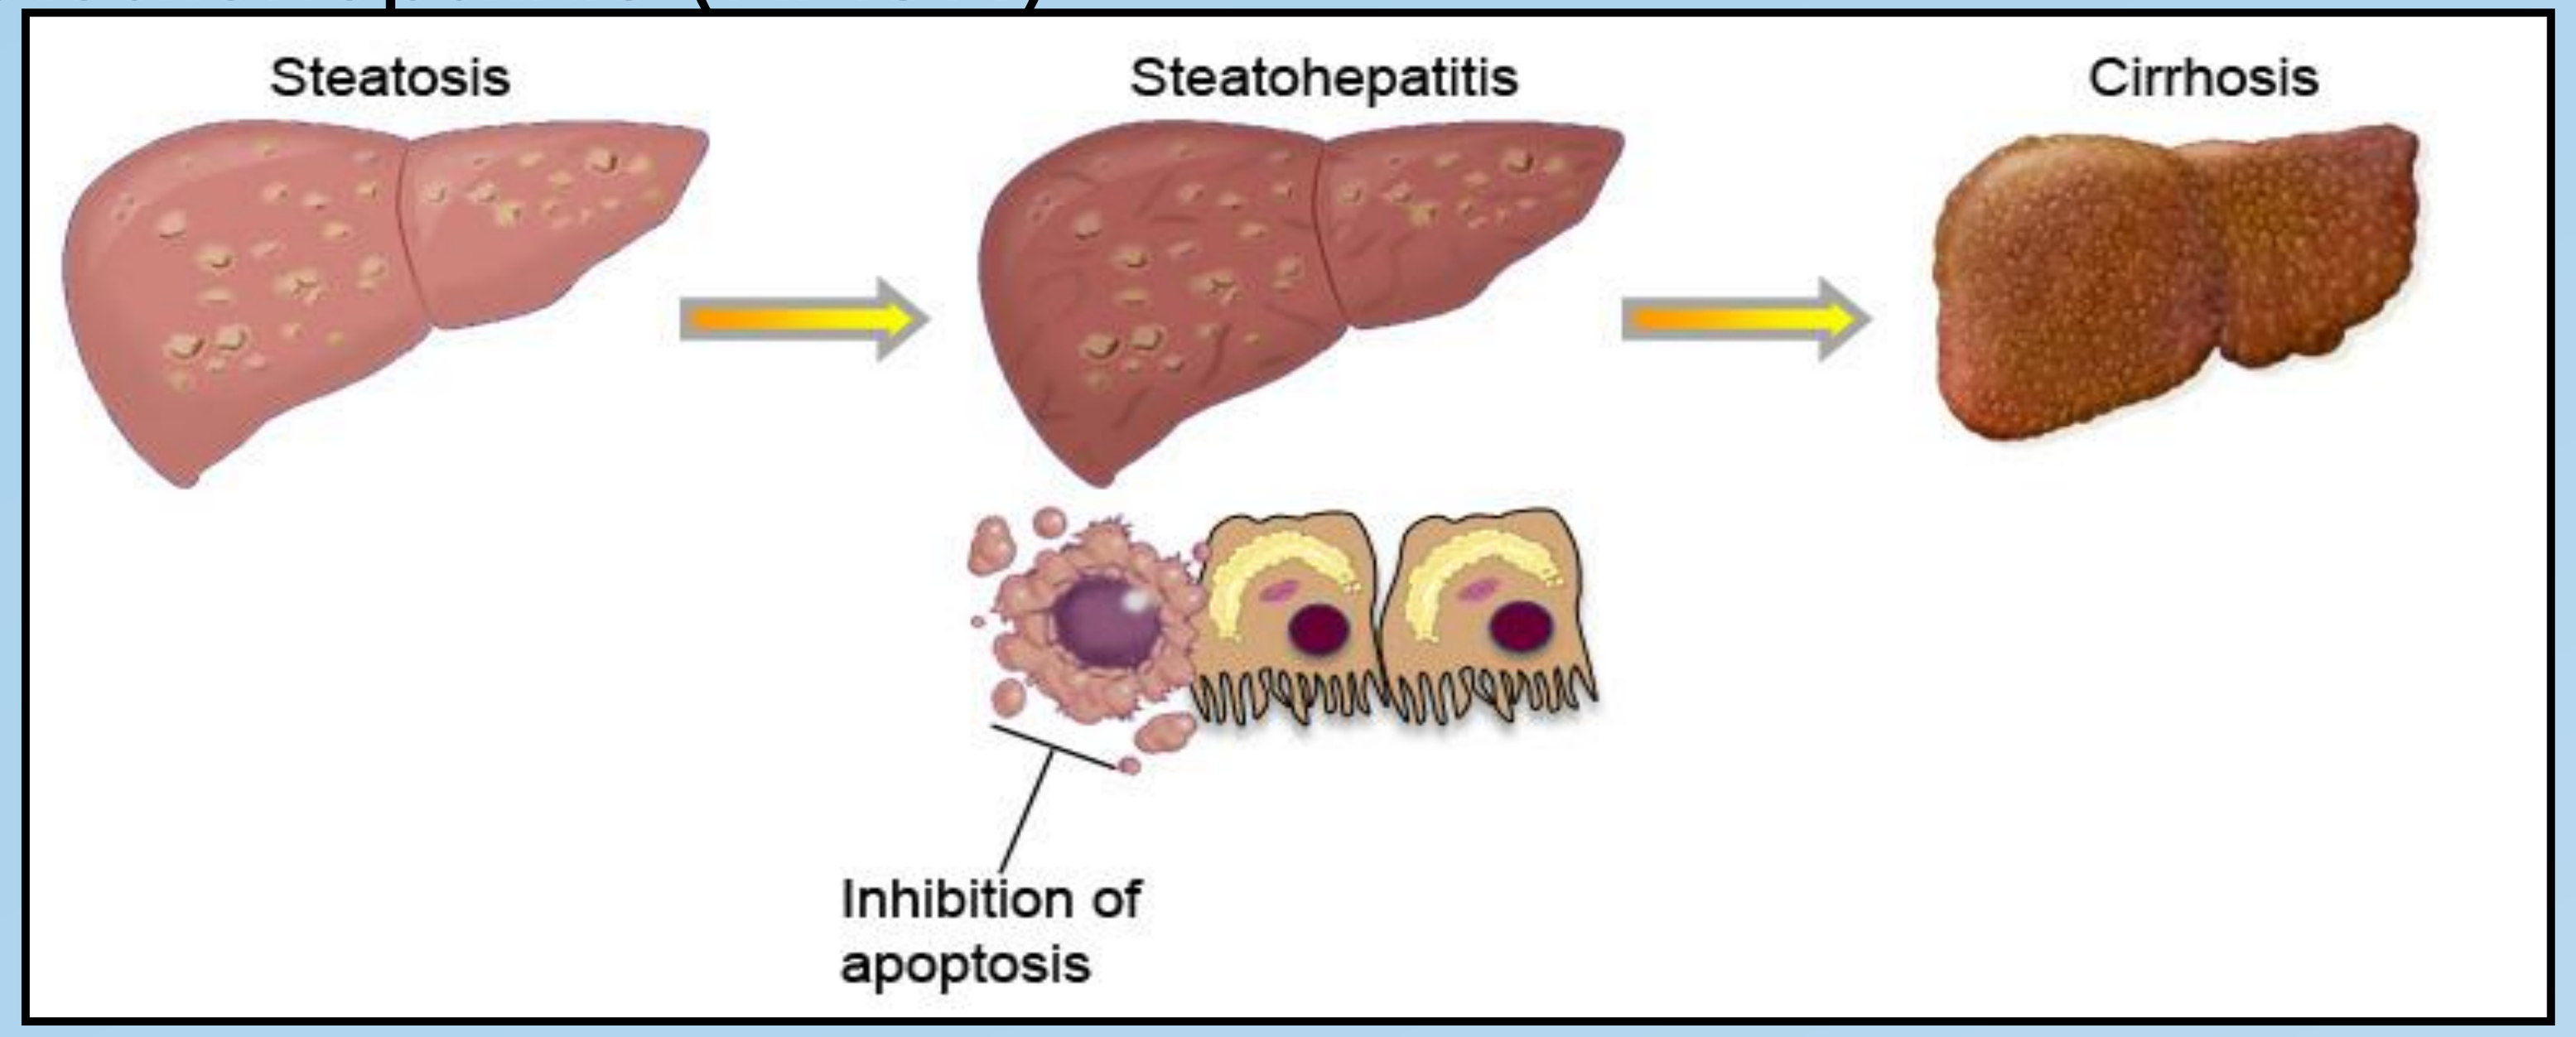
Steatosis
Steatohepatitis
Cirrhosis
Inhibition of apoptosis

➢Emerging data suggests that inhibition of **caspases** may be an attractive therapeutic approach for patients with NASH

➢Our **aim** was to determine if the pan-caspase inhibitor, IDN-6556, reduces hepatocellular apoptosis and fibrosis

METHODS

- C57BL/6 mice were fed a CDAA or a choline-sufficient amino acid-defined (CSAA) diet for 20 weeks
- Starting on week 16, mice fed the CDAA diet were subject to drug or placebo treatment via gavage for five weeks

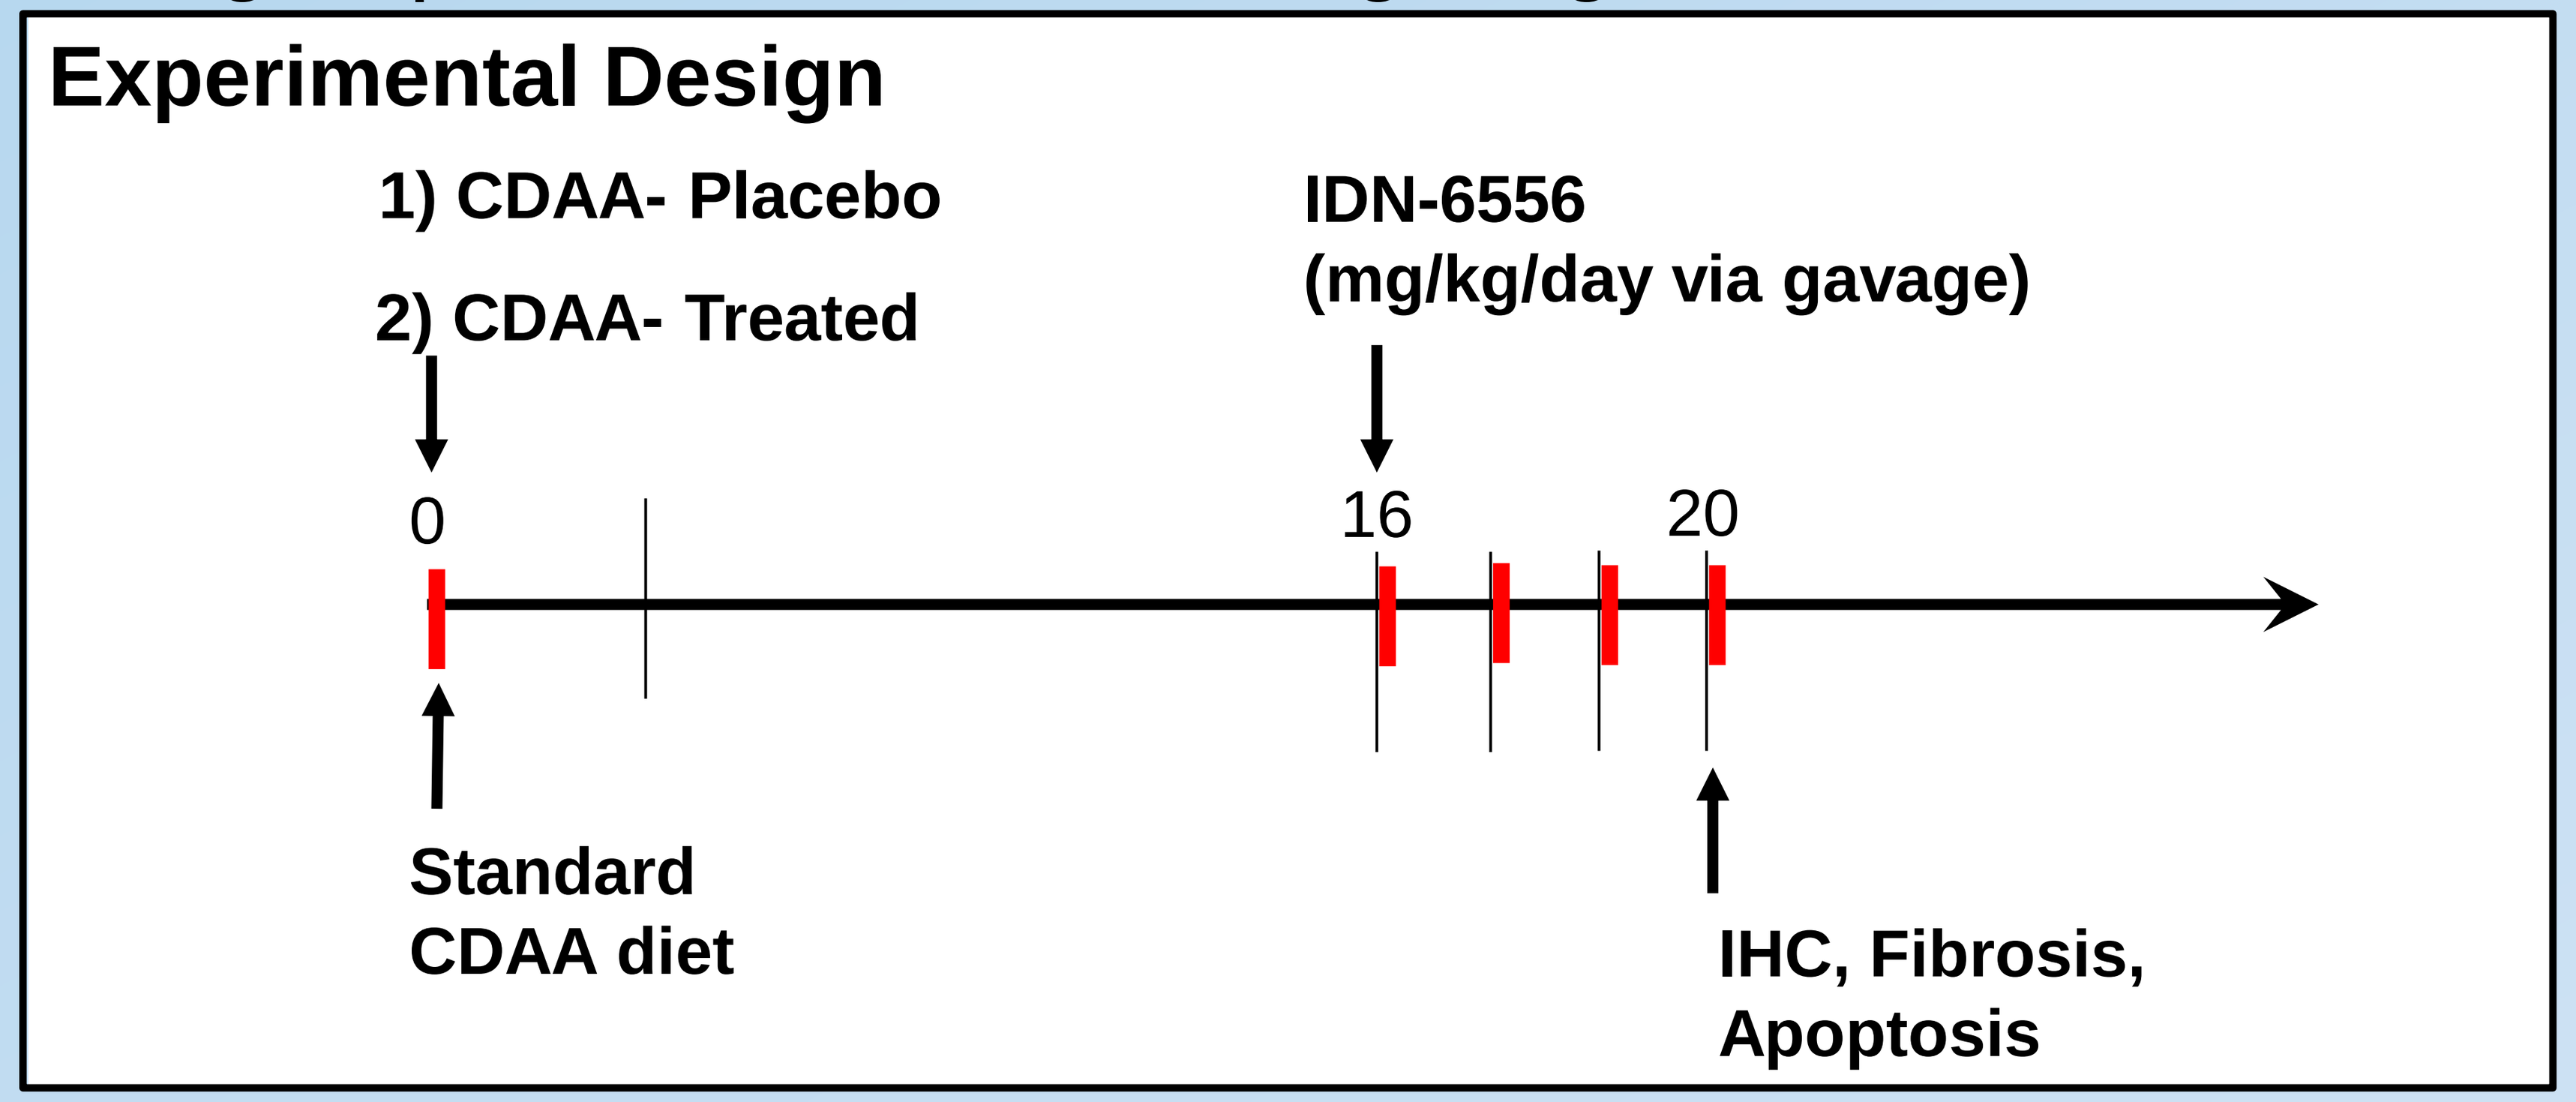
Experimental Design
1) CDAA- Placebo
2) CDAA- Treated
IDN-6556
(mg/kg/day via gavage)
0
16
20
Standard
CDAA diet
IHC, Fibrosis,
Apoptosis

- Liver and blood were collected after week 20
- Hepatocellular apoptosis, fibrosis and inflammatory activity were assessed

RESULTS

Liver Fibrosis was diminished in treated mice: Sirius Red quantification and expression of CTGF mRNA

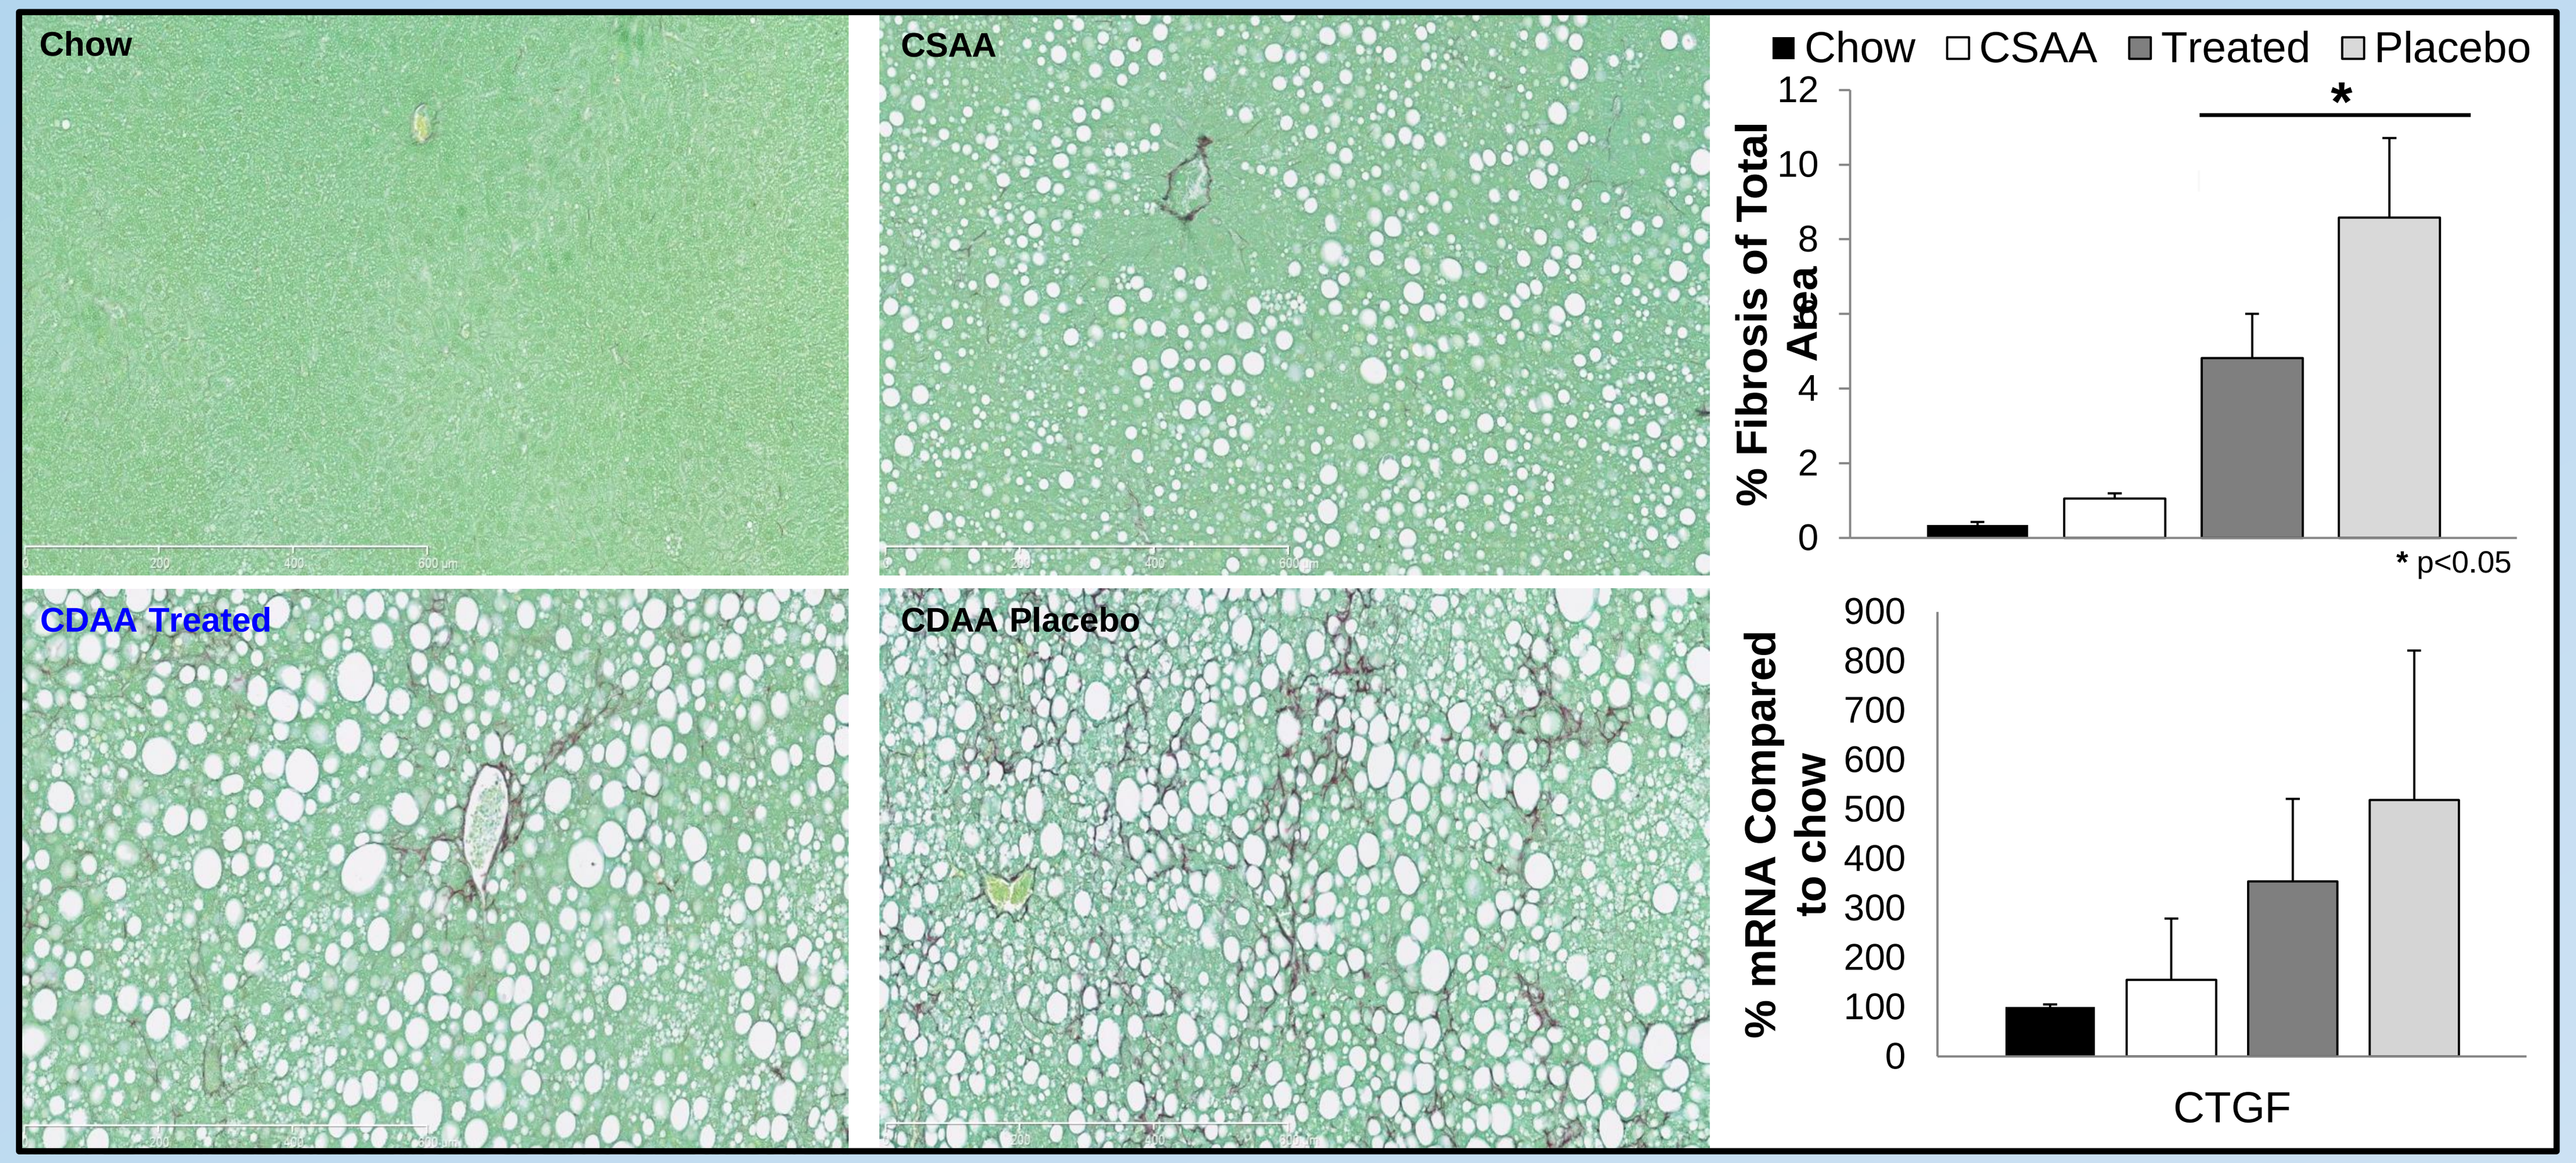
Chow
CSAA
CDAA Treated
CDAA Placebo
Chow CSAA Treated Placebo
% Fibrosis of Total Area
* p<0.05
% mRNA Compared to chow
CTGF

IDN-6556 administration reduced Hepatic Stellate Cell (HSC) activation: Real-time quantitative PCR of fibrogenic genes in isolated stellate cells

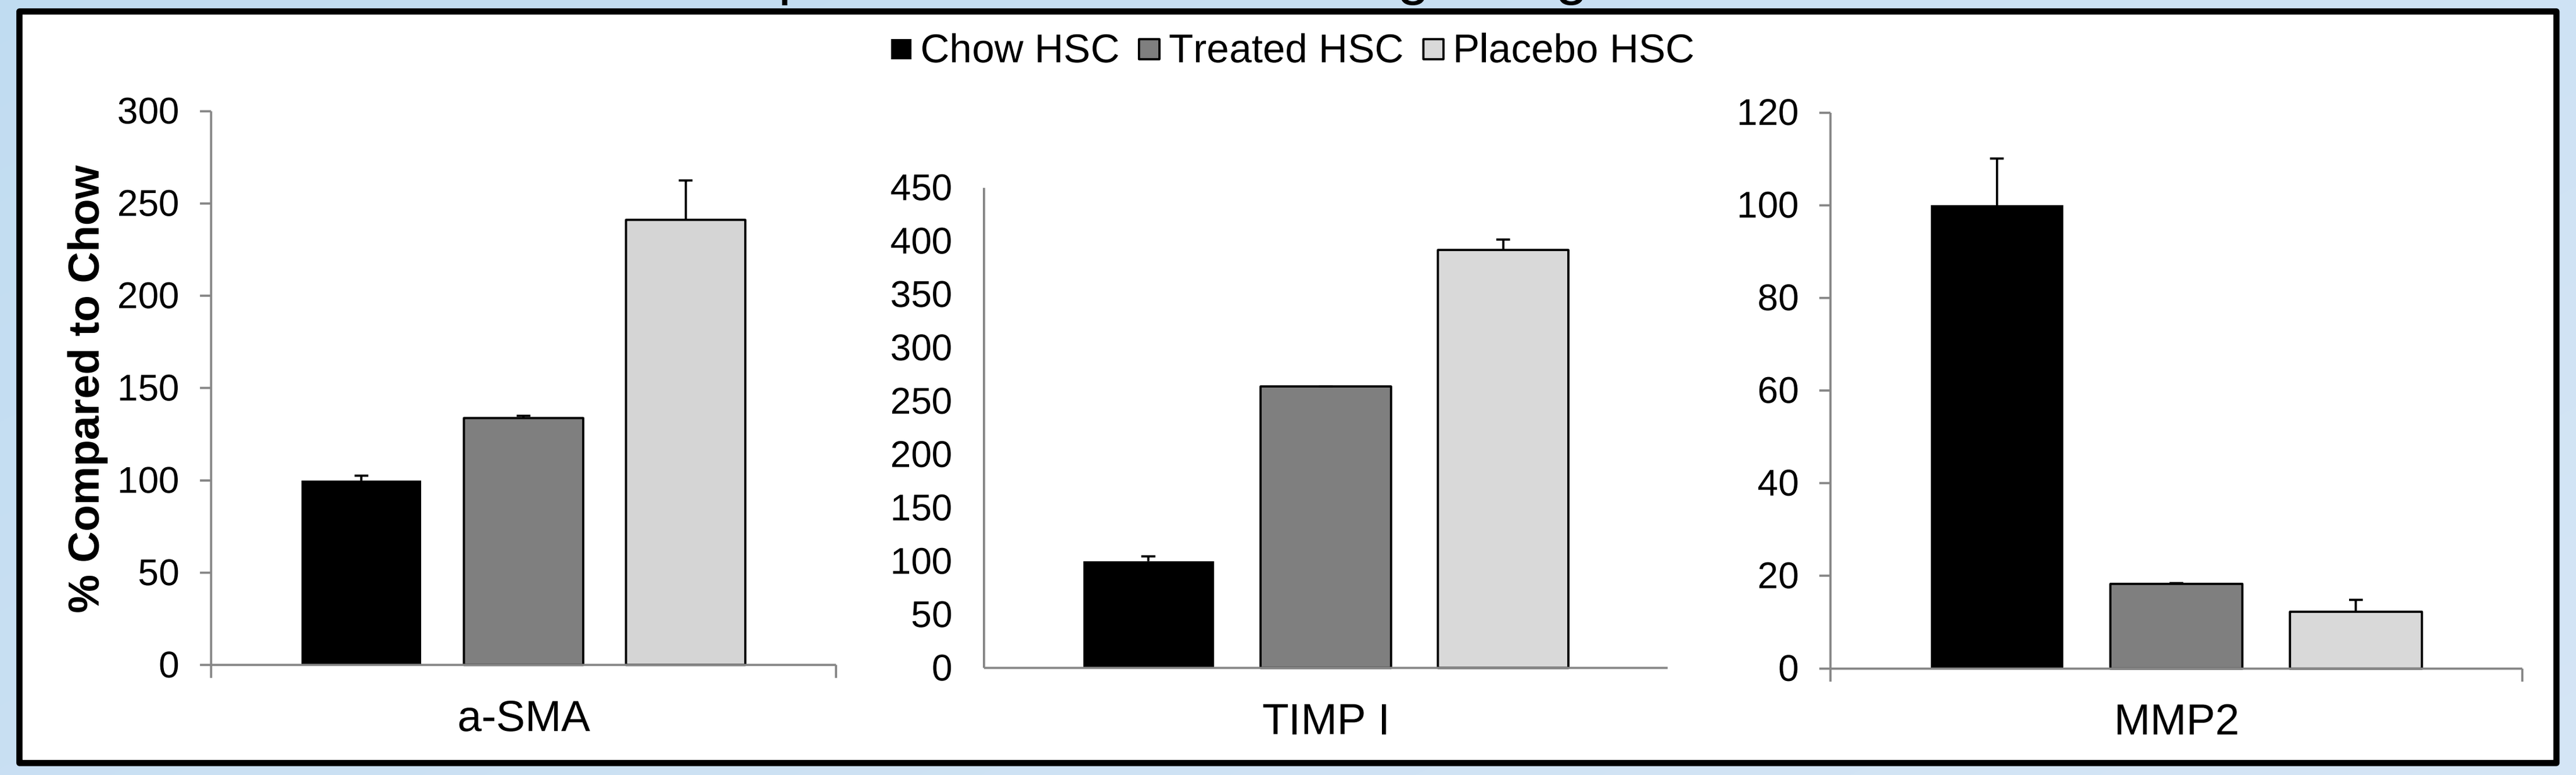
Chow HSC Treated HSC Placebo HSC
% Compared to Chow
a-SMA
TIMP I
MMP2

Mice that received the pan-caspase inhibitor showed inhibition of apoptosis: Bid, Cleaved Caspase 3 and Cytokeratin 18 Immunoblots

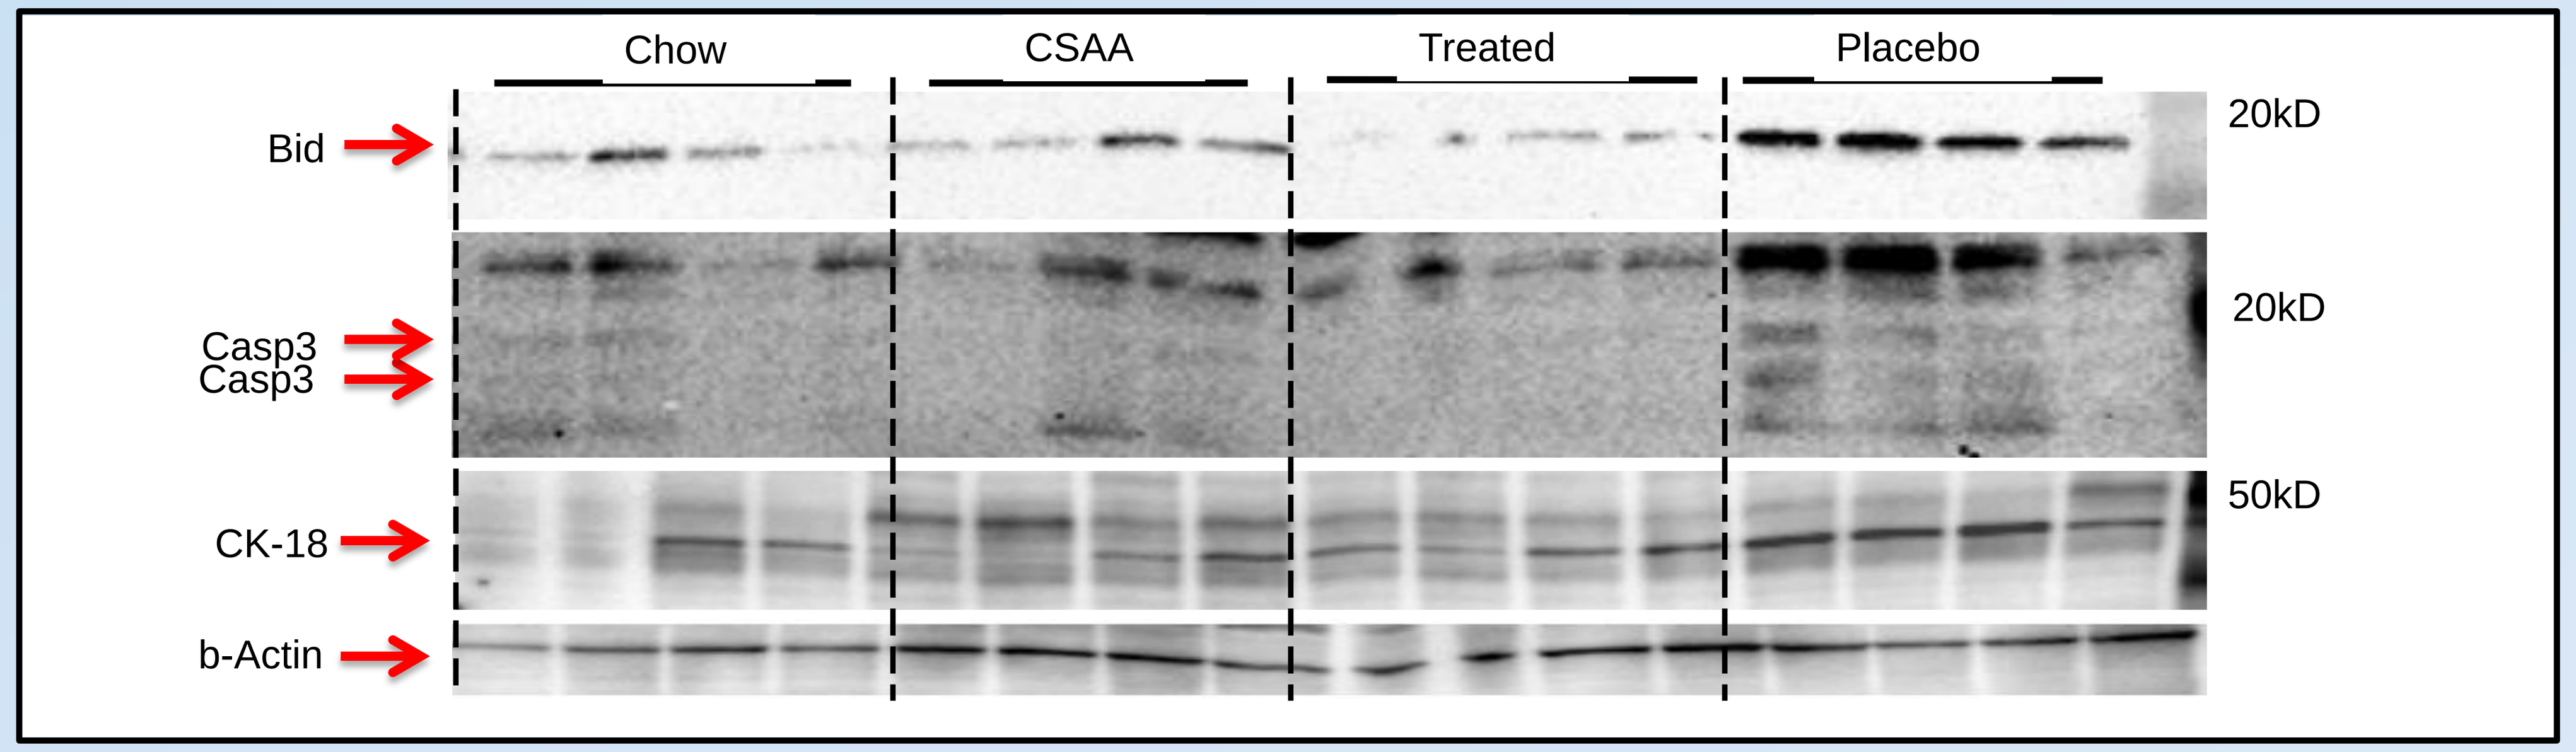
Chow
CSAA
Treated
Placebo
Bid
Casp3
Casp3
CK-18
b-Actin
20kD
20kD
50kD

RESULTS

Neutrophil infiltration in the liver declined after exposure to the drug: Myeloperoxidase (MPO) Staining and Osteopontin mRNA Expression

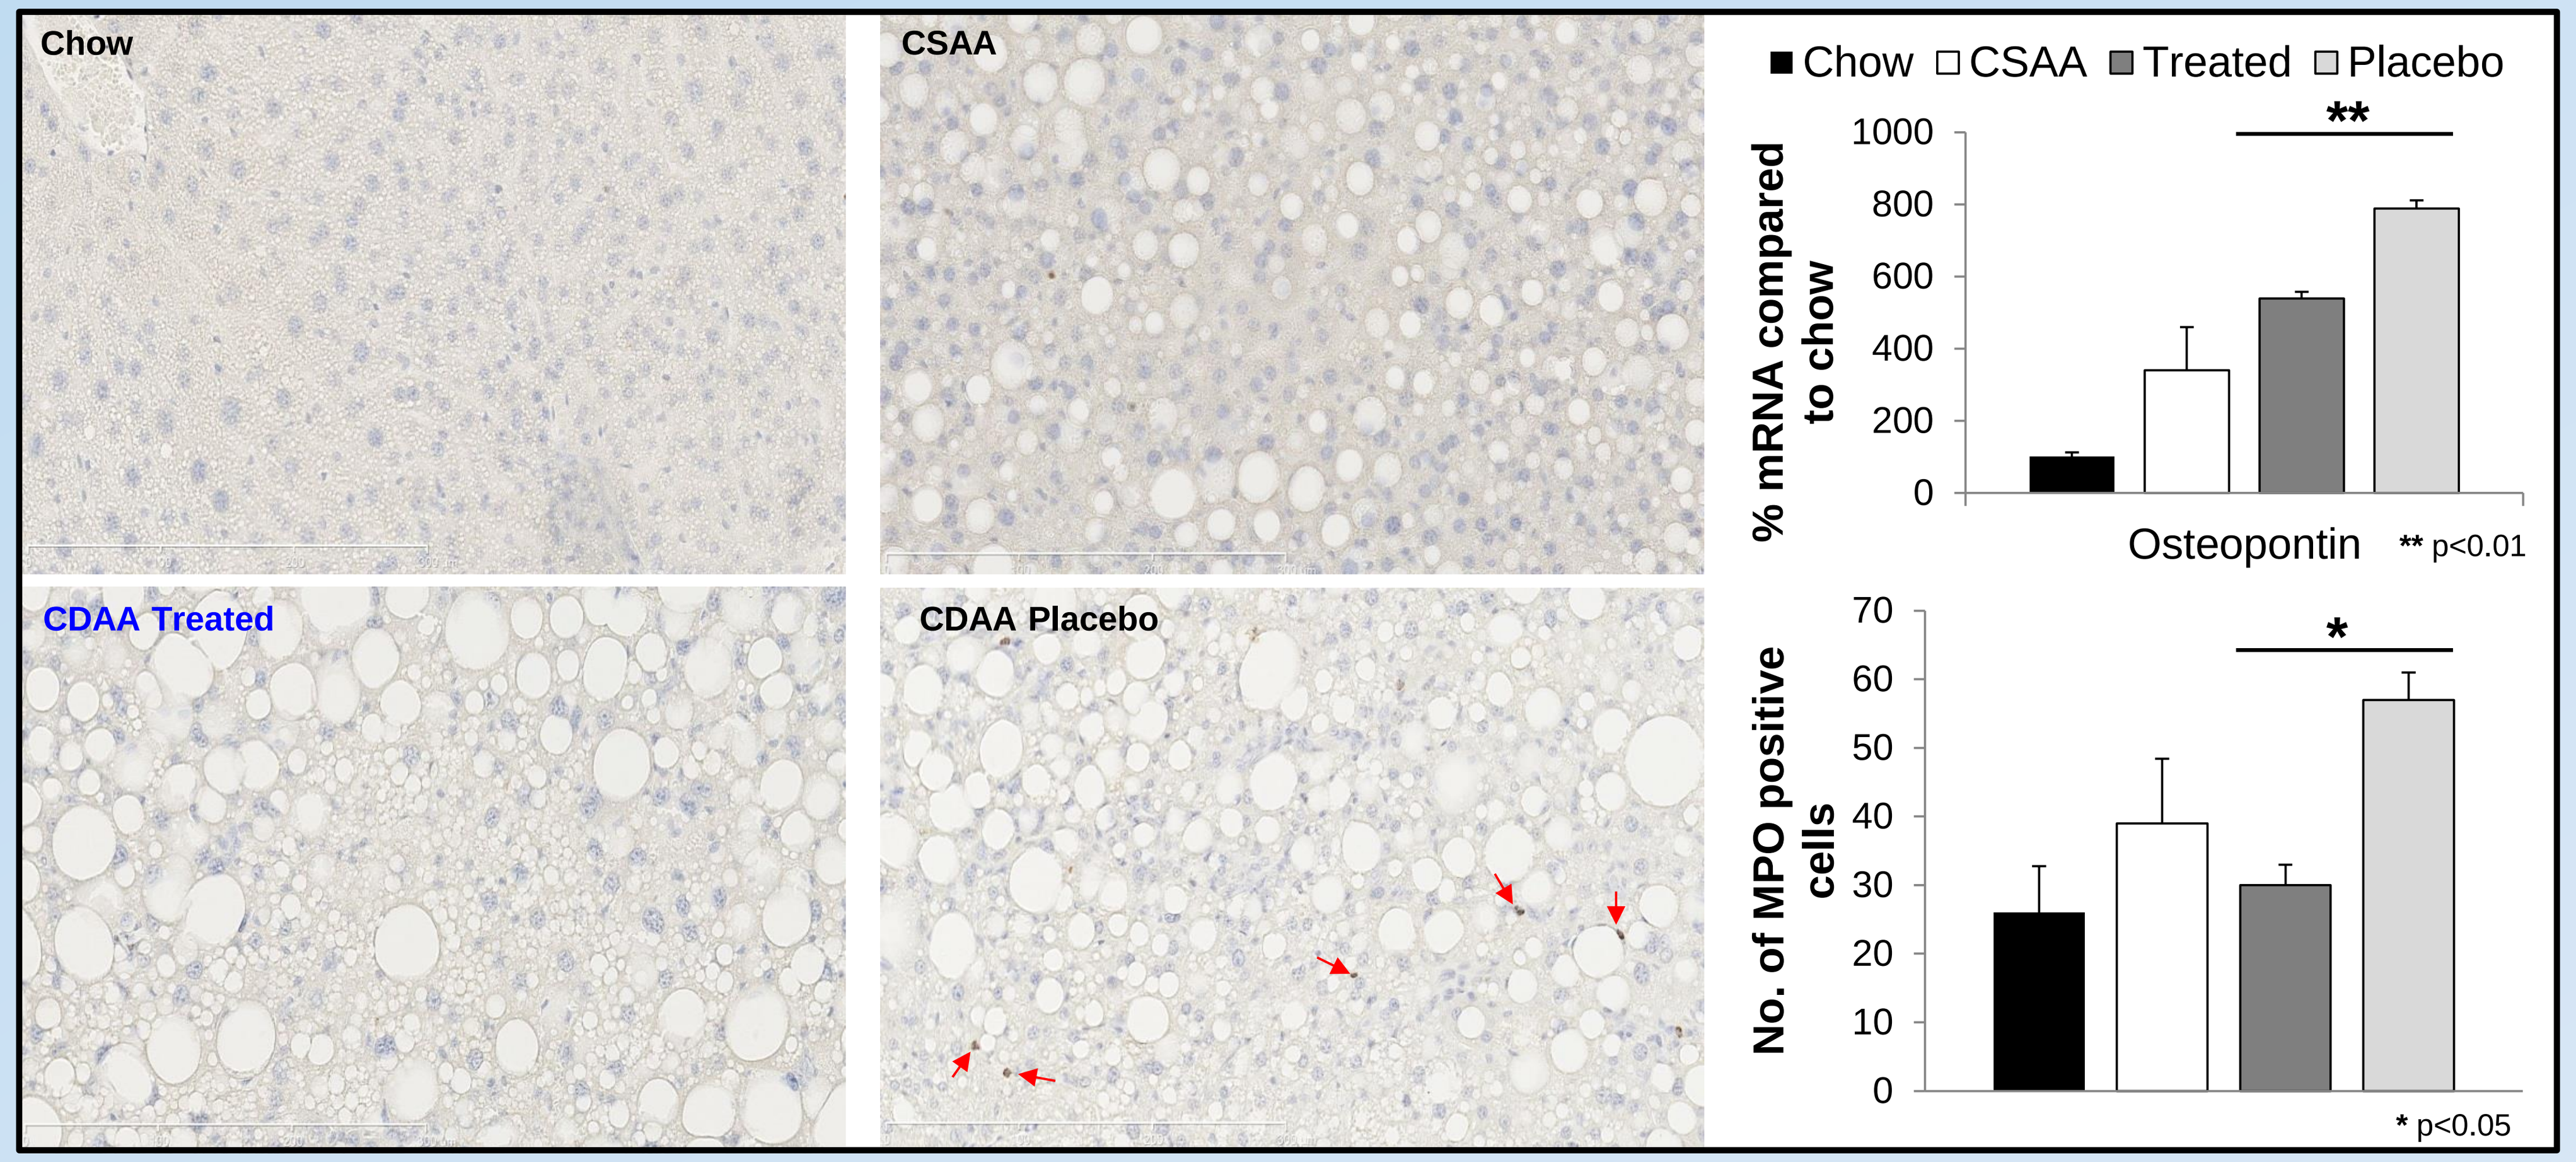
Chow
CSAA
CDAA Treated
CDAA Placebo
Chow CSAA Treated Placebo
% mRNA compared to chow
Osteopontin
** p<0.01
No. of MPO positive cells
* p<0.05

CONCLUSION

The data shown suggests that oral administration of the experimental drug IDN-6556 inhibits apoptosis and attenuates fibrosis and inflammation associated with experimental NASH. ALT/AST levels (not shown) showed no marked decrease between CDAA treated and CDAA placebo groups, while Insulin levels (not shown) showed a significant decrease in the treated group. The findings indicate that IDN-6556 might be a promising method of therapy and warrants further research

FUNDING

This study was supported in part by an unrestricted grant from Conatus Pharmaceutical and NIH grant DK082451 to AEF

EASL THE INTERNATIONAL LIVER CONGRESS™ 2013
48th ANNUAL MEETING OF THE EUROPEAN ASSOCIATION FOR THE STUDY OF THE LIVER
AMSTERDAM, THE NETHERLANDS, APRIL 24-28 / 2013